The Aldehyde Trap NS2 Reduces Ocular Inflammation in an Endotoxin-Induced Model in Rats

Kenneth J. Mandell, Scott L. Young, Todd C. Brady; Aldeyra Therapeutics, Lexington, MA
kmandell@aldeyra.com

Presentation #3095

Abstract

Purpose: NS2 is an investigational drug in development for acute noninfectious anterior uveitis and other ocular indications. The mechanism of action of NS2 involves neutralizing toxic free-aldehydes, which are mediators of inflammatory disease, both in the eye and elsewhere. The purpose of this study was to evaluate efficacy of a topical NS2 eye drop formulation in a rat model of ocular inflammation.

Methods: Ocular inflammation was induced in Lewis rats by foot pad injection of lipopolysaccharide (LPS). Thirty (30) female Lewis rats were split in 3 groups to receive topical NS2 0.5%, balanced salt solution (BSS) or dexamethasone 0.1% (DEX) at 1, 3, 7, 10 and 17 hours post-induction. An additional 2 groups received a single invtravitreal dose NS2 or BSS at the time of induction. Ocular exams were performed at 24 hours post-induction and scored using a combined Combined Draize and McDonald-Shadduck scoring system. Animals were euthanized to collect ocular tissues, and levels of inflammatory markers ICAM-1 (Intercellular Adhesion Molecule 1), IL-6 (Interleukin 6) and MCP-1 (Monocyte Chemoattractant Protein 1) were measured from retina-choroid specimens by enzyme-linked immunosorbent assay (ELISA).

Results: The mean total ocular examination (OE) scores at 24 hours post-LPS induction were 21.4, 10.1 and 15.7 for the topical BSS, DEX and NS2 groups, respectively. These reductions in mean total OE scores were statistically significant for both NS2 and DEX relative to the BSS control. Similarly, scores for individual components of the OE revealed similar trends with statistically significant reductions observed for conjunctival hyperemia, iris hyperemia and anterior chamber flare for both NS2 and DEX. Statistically significant reductions in ICAM-1 levels were observed for both NS2 and DEX relative to BSS. However, a statistically significant reduction in IL-6 was observed only for NS2. MCP-1 levels could not be detected in any group. Intravitreal NS2 showed similar trends in reduction of clinical scores relative to BSS, but there were no statistically significant differences in cytokine levels detected.

Conclusions:Together these findings provide symptomatic and biochemical evidence of the anti-inflammatory effects of the aldehyde trap NS2 in a model of inflammatory eye disease. The production of toxic aldehydes is mechanistically linked to uveitis and a variety of other inflammatory ocular diseases. The ability of NS2 to trap and sequester free-aldehydes may prove to be a novel mechanism for the treatment of ocular inflammatory diseases.

Disclosure: Kenneth J. Mandell, Aldeyra Therapeutics (Code C (Consultant)); Scott L. Young, Aldeyra Therapeutics (Code E (Employment)); Todd C. Brady, Aldeyra Therapeutics (Code E (Employment))

NS2 Reduces Ocular Inflammation

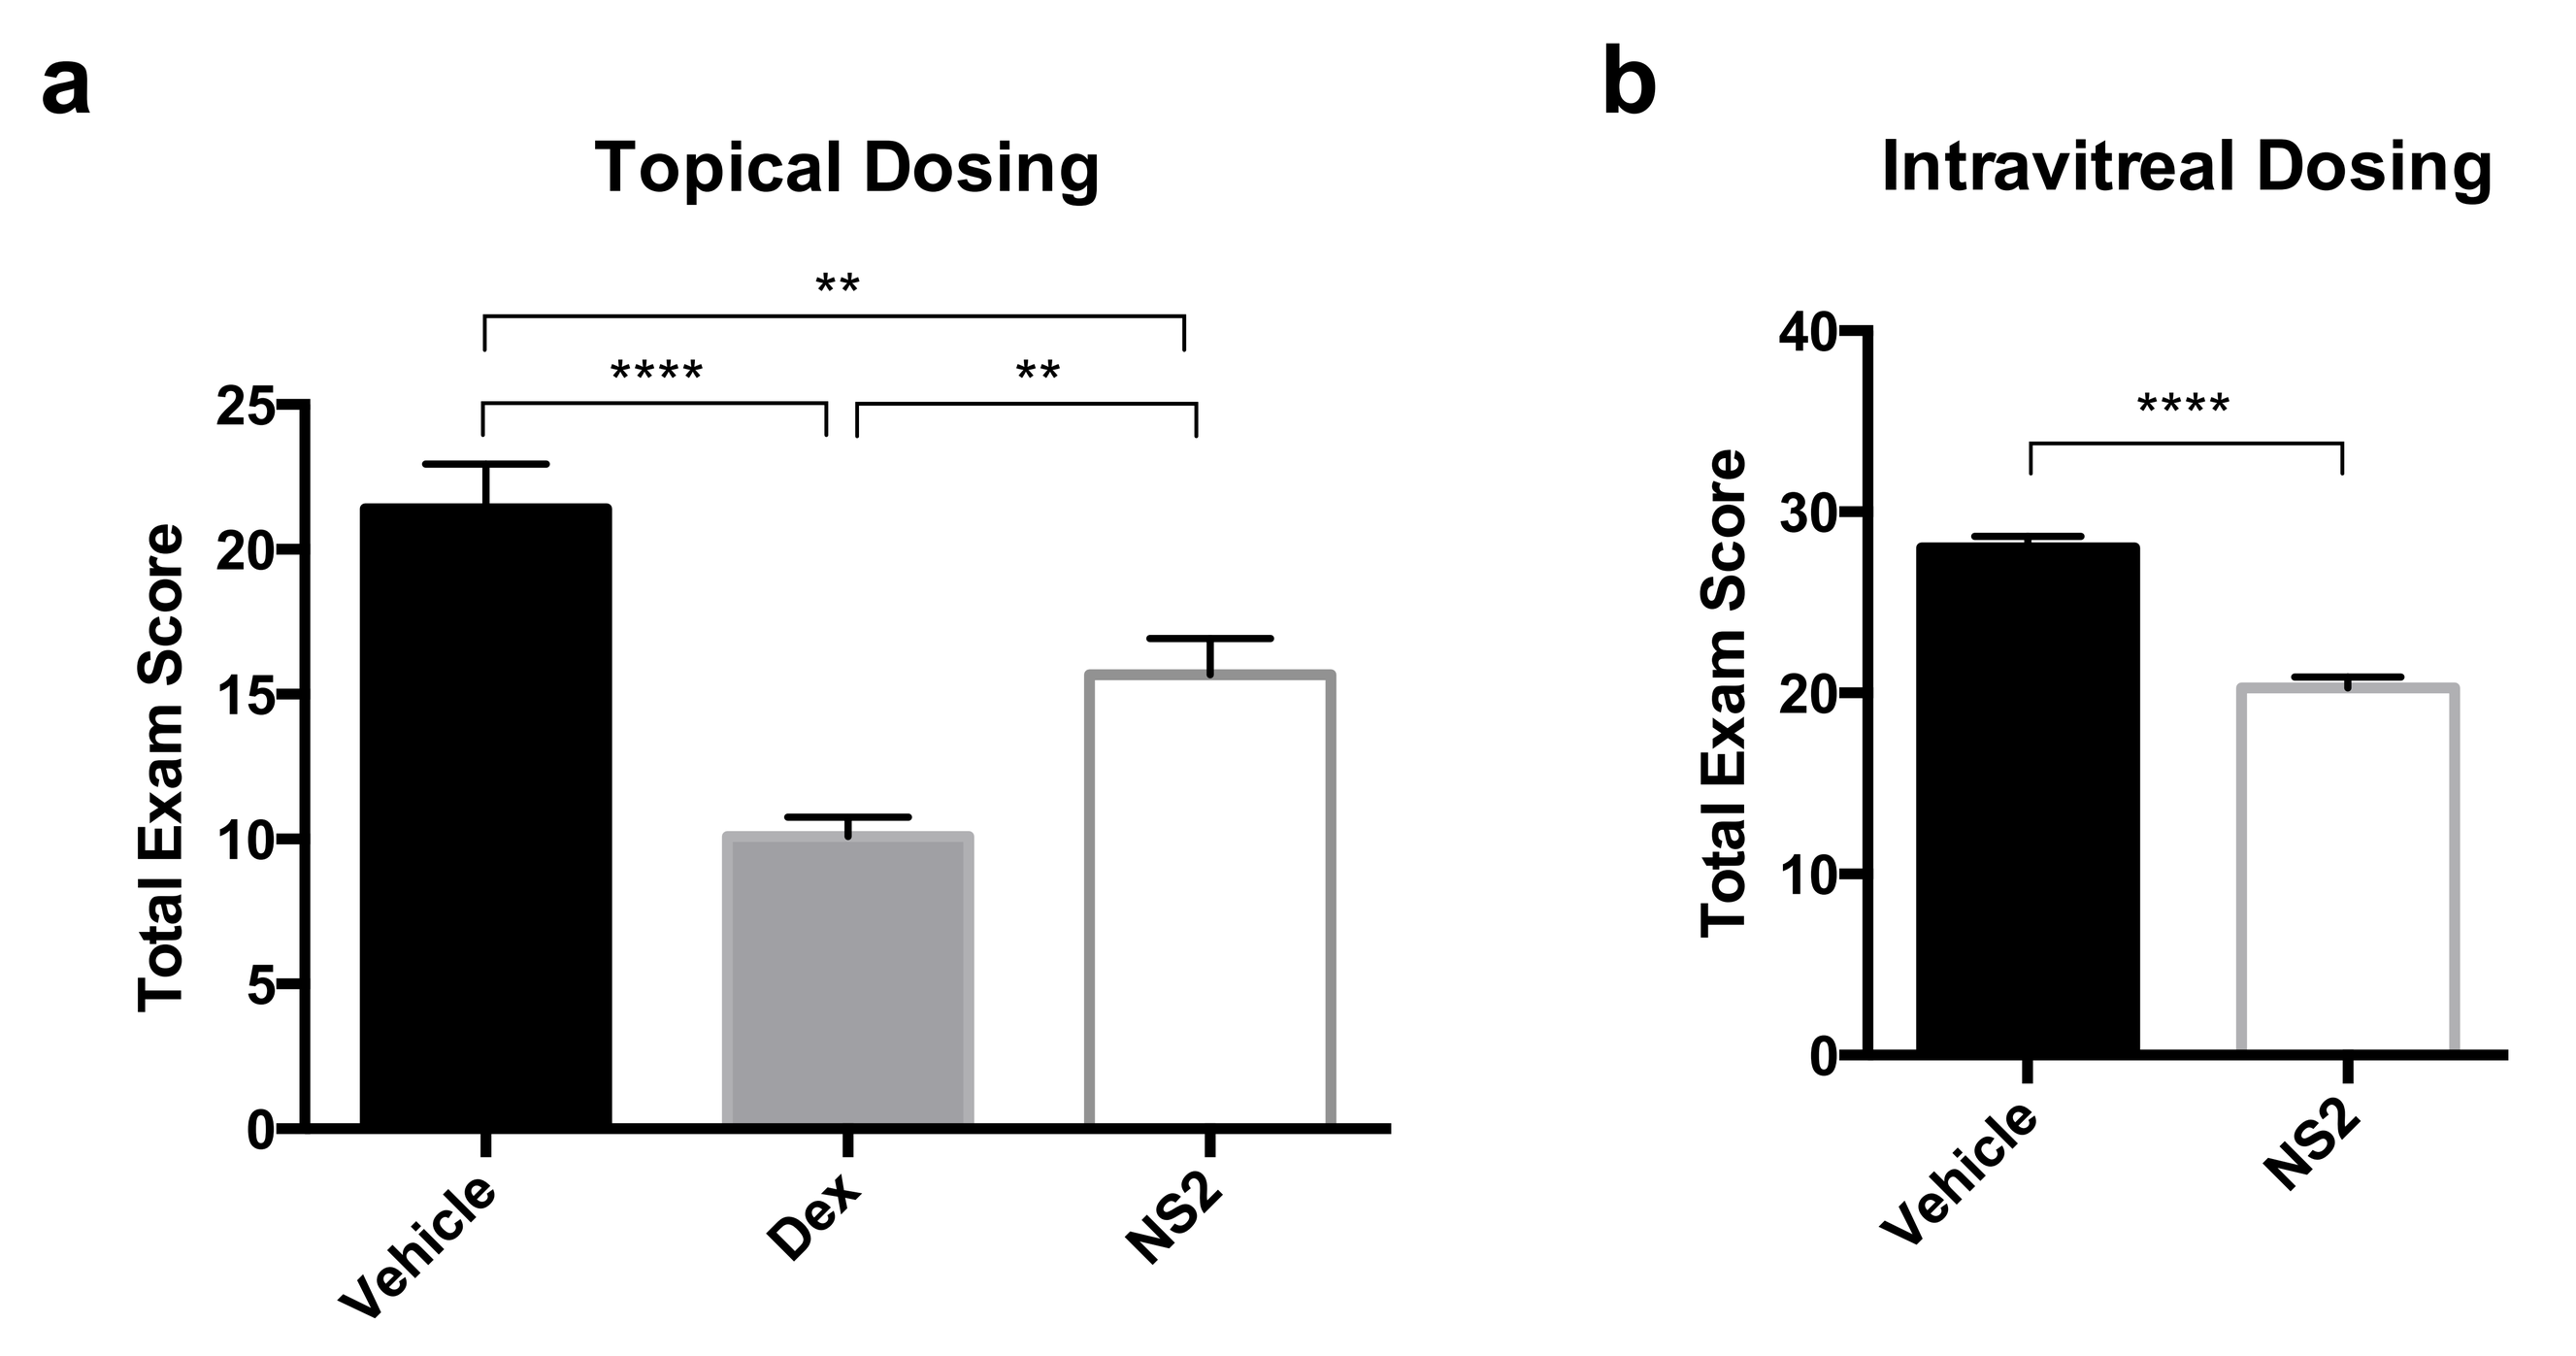
a
Topical Dosing
Total Exam Score
25
20
15
10
5
0

**
**
Vehicle
Dex
NS2
b
Intravitreal Dosing
Total Exam Score
40
30
20
10
0

Vehicle
NS2

Figure 1. Total Ocular Exam Scores. Total exam scores for animals (a) dosed topically with vehicle, dexamethasone or NS2; or (b) dosed intravitreally with vehicle or NS2. * p < 0.05, ** p < 0.01, *** p < 0.001, **** p < 0.0001.

NS2 Reduces Conjunctival and Iris Hyperemia and AC Flare

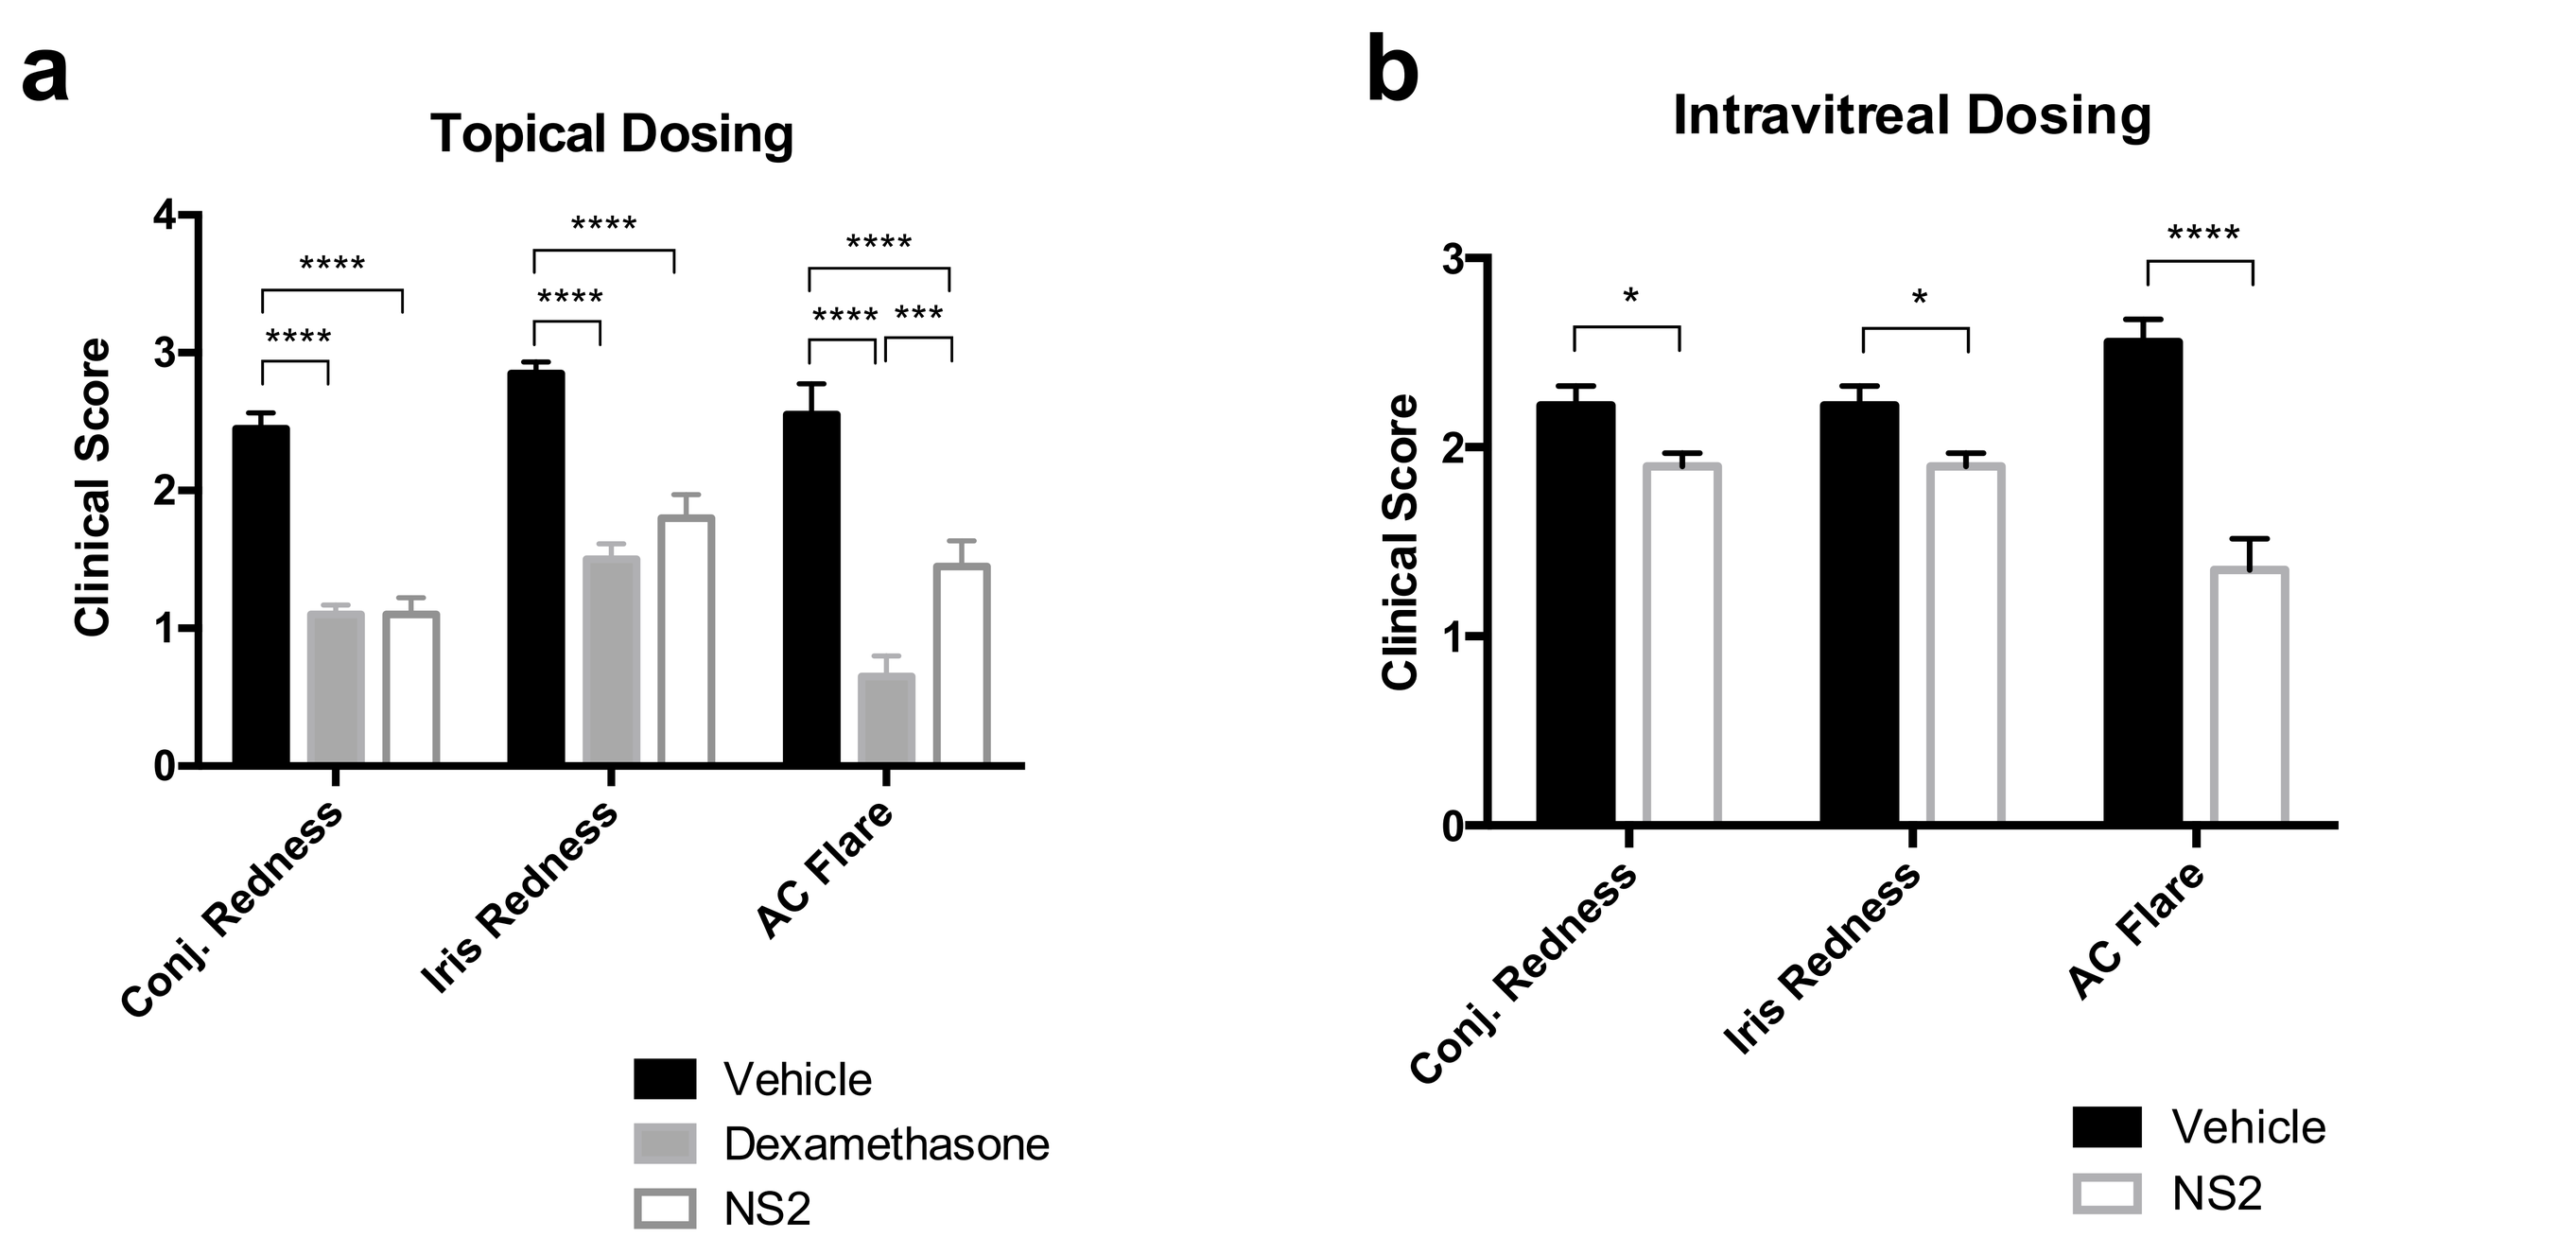
a
Topical Dosing
Clinical Score
Conj. Redness
Iris Redness
AC Flare
Vehicle
Dexamethasone
NS2
b
Intravitreal Dosing
Clinical Score
Conj. Redness
Iris Redness
AC Flare
Vehicle
NS2

Figure 2. Individual Exam Scores. Individual exam scores for conjunctival redness, iris redness and anterior chamber flare in animals (a) dosed topically with vehicle, dexamethasone or NS2; or (b) dosed intravitreally with vehicle or NS2. * p < 0.05, ** p < 0.01, *** p < 0.001, **** p < 0.0001.

Anterior Chamber Cell and Protein Levels

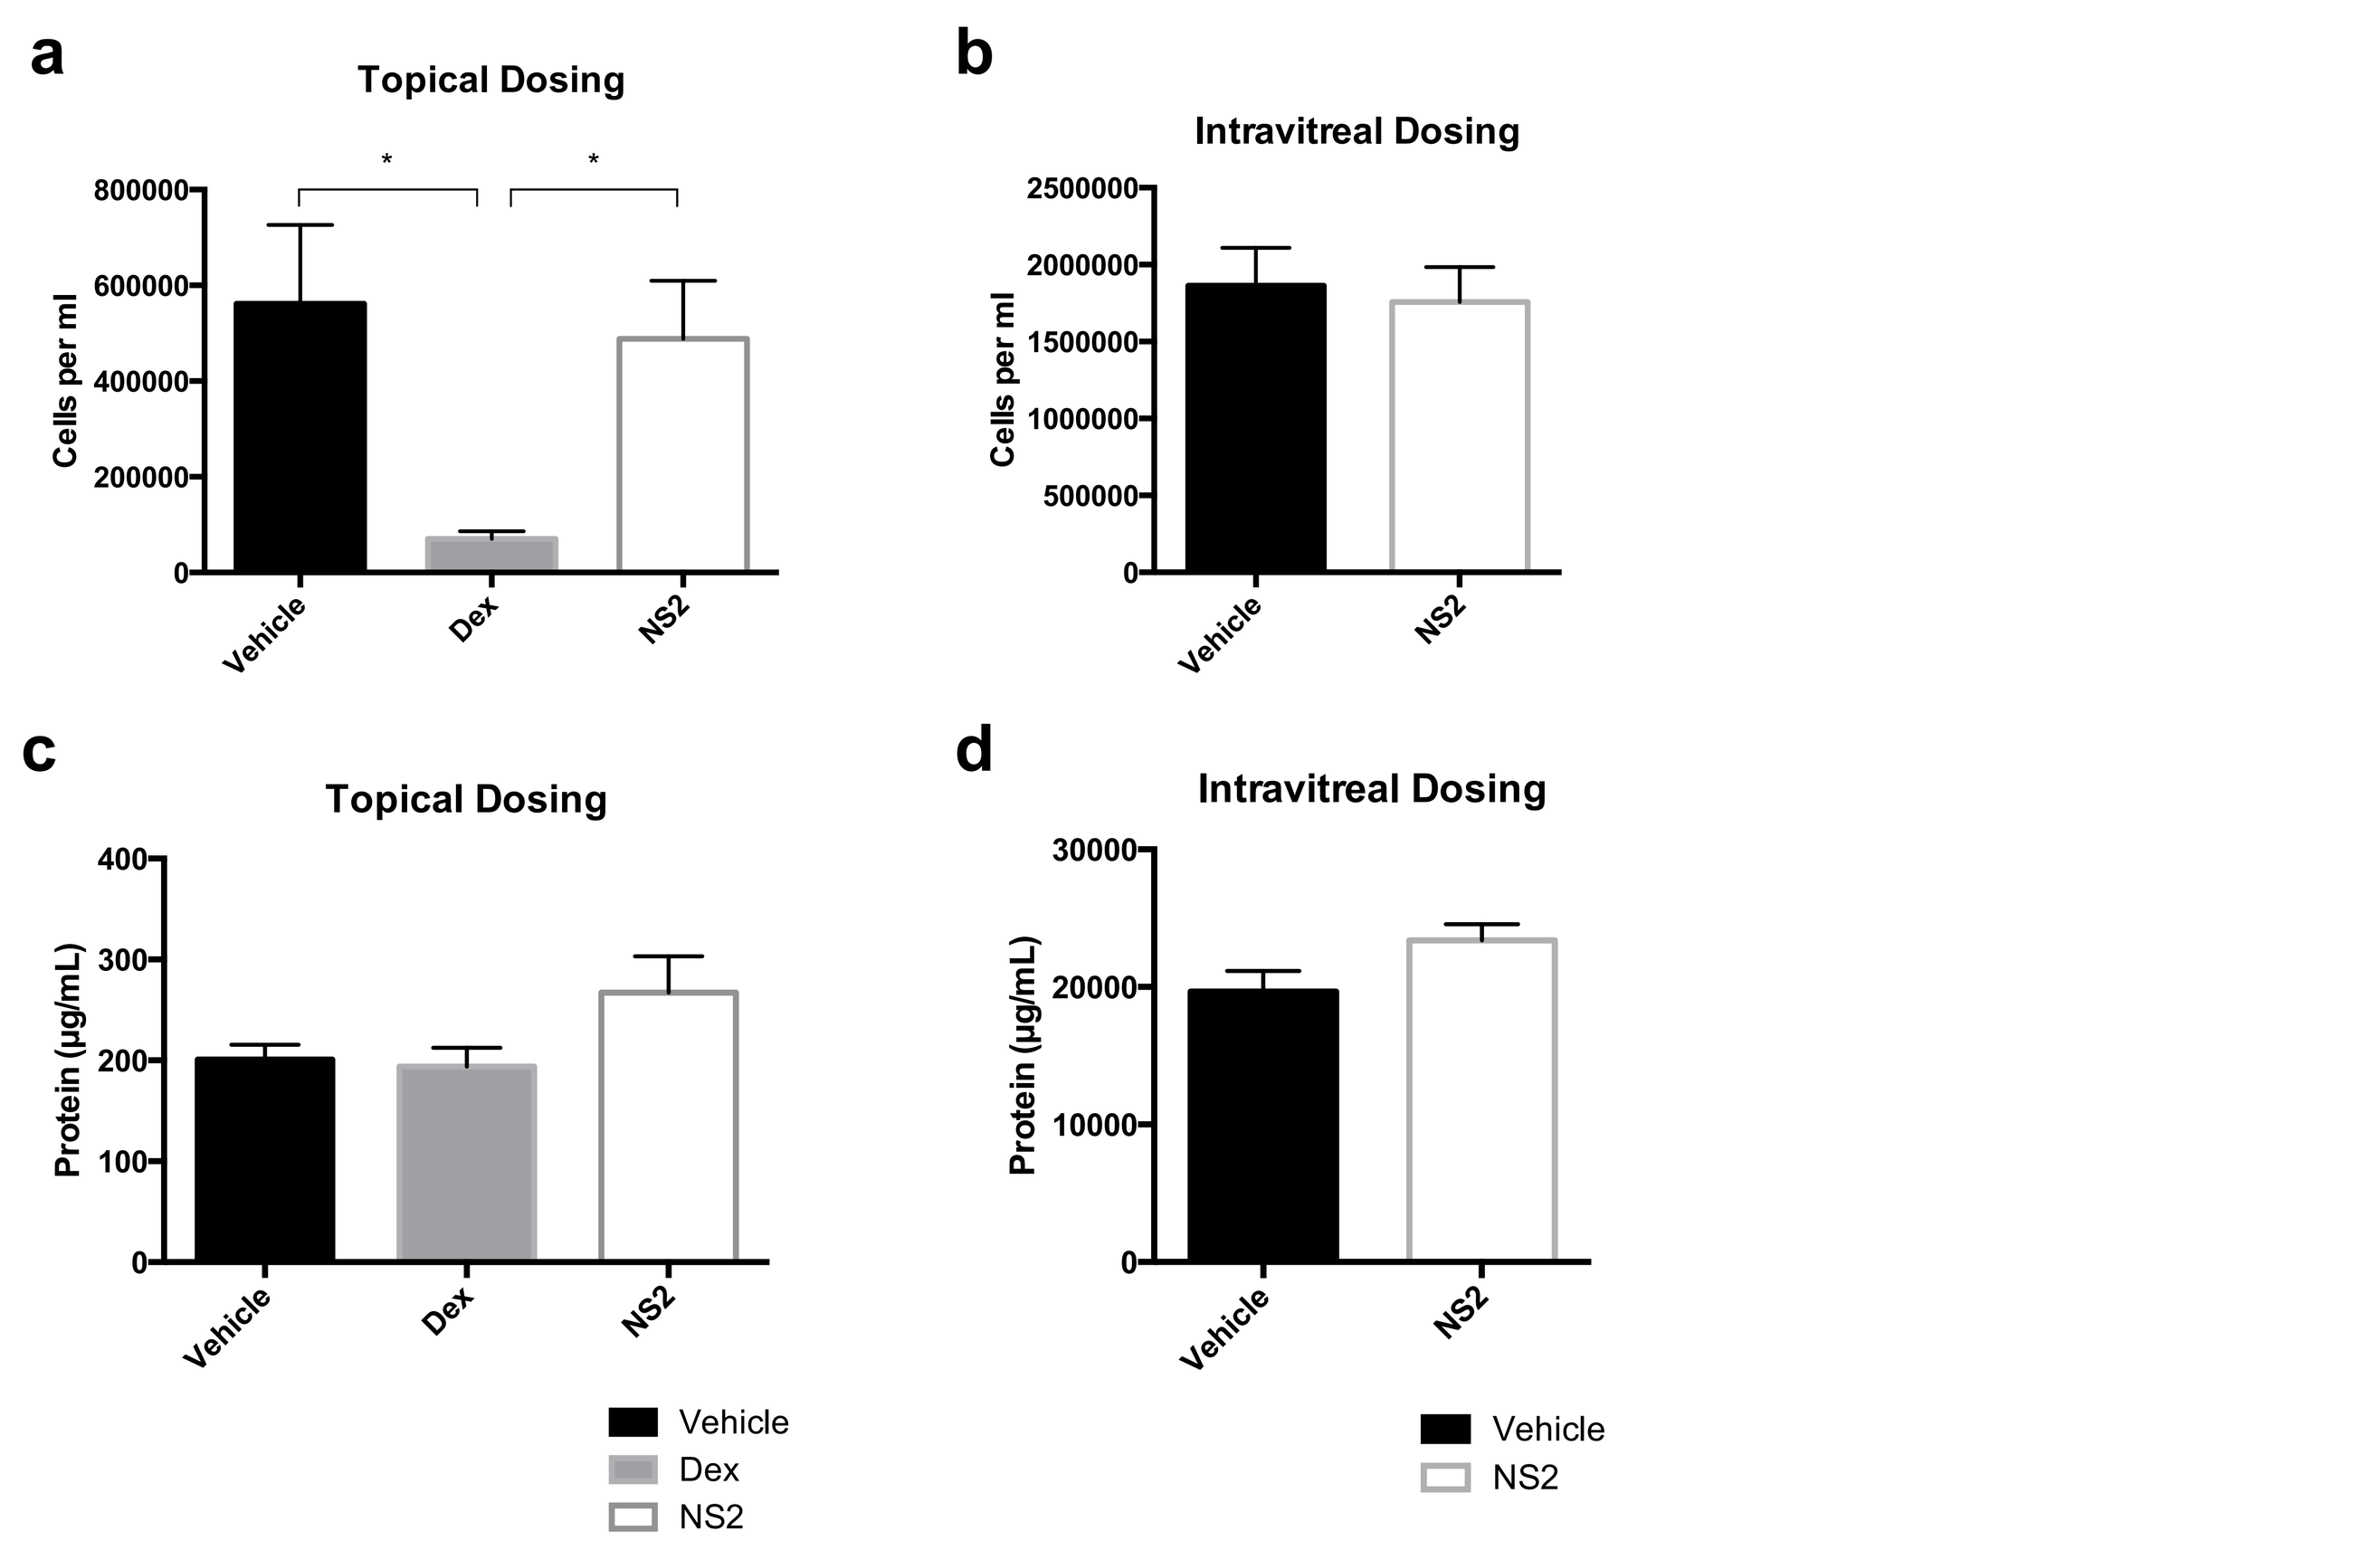
a
Topical Dosing
Cells per ml
Vehicle
Dex
NS2
b
Intravitreal Dosing
Cells per ml
Vehicle
NS2
c
Topical Dosing
Protein (µg/mL)
Vehicle
Dex
NS2
d
Intravitreal Dosing
Protein (µg/mL)
Vehicle
NS2

Figure 3. Anterior Chamber Cell and Protein Levels. Anterior chamber cell counts and protein levels from animals dosed topically with vehicle, dexamethasone or NS2 (a,c); or dosed intravitreally with vehicle or NS2 (b,d). * p < 0.05, ** p < 0.01, *** p < 0.001, **** p < 0.0001.

NS2 Reduces IL-6 and ICAM-1

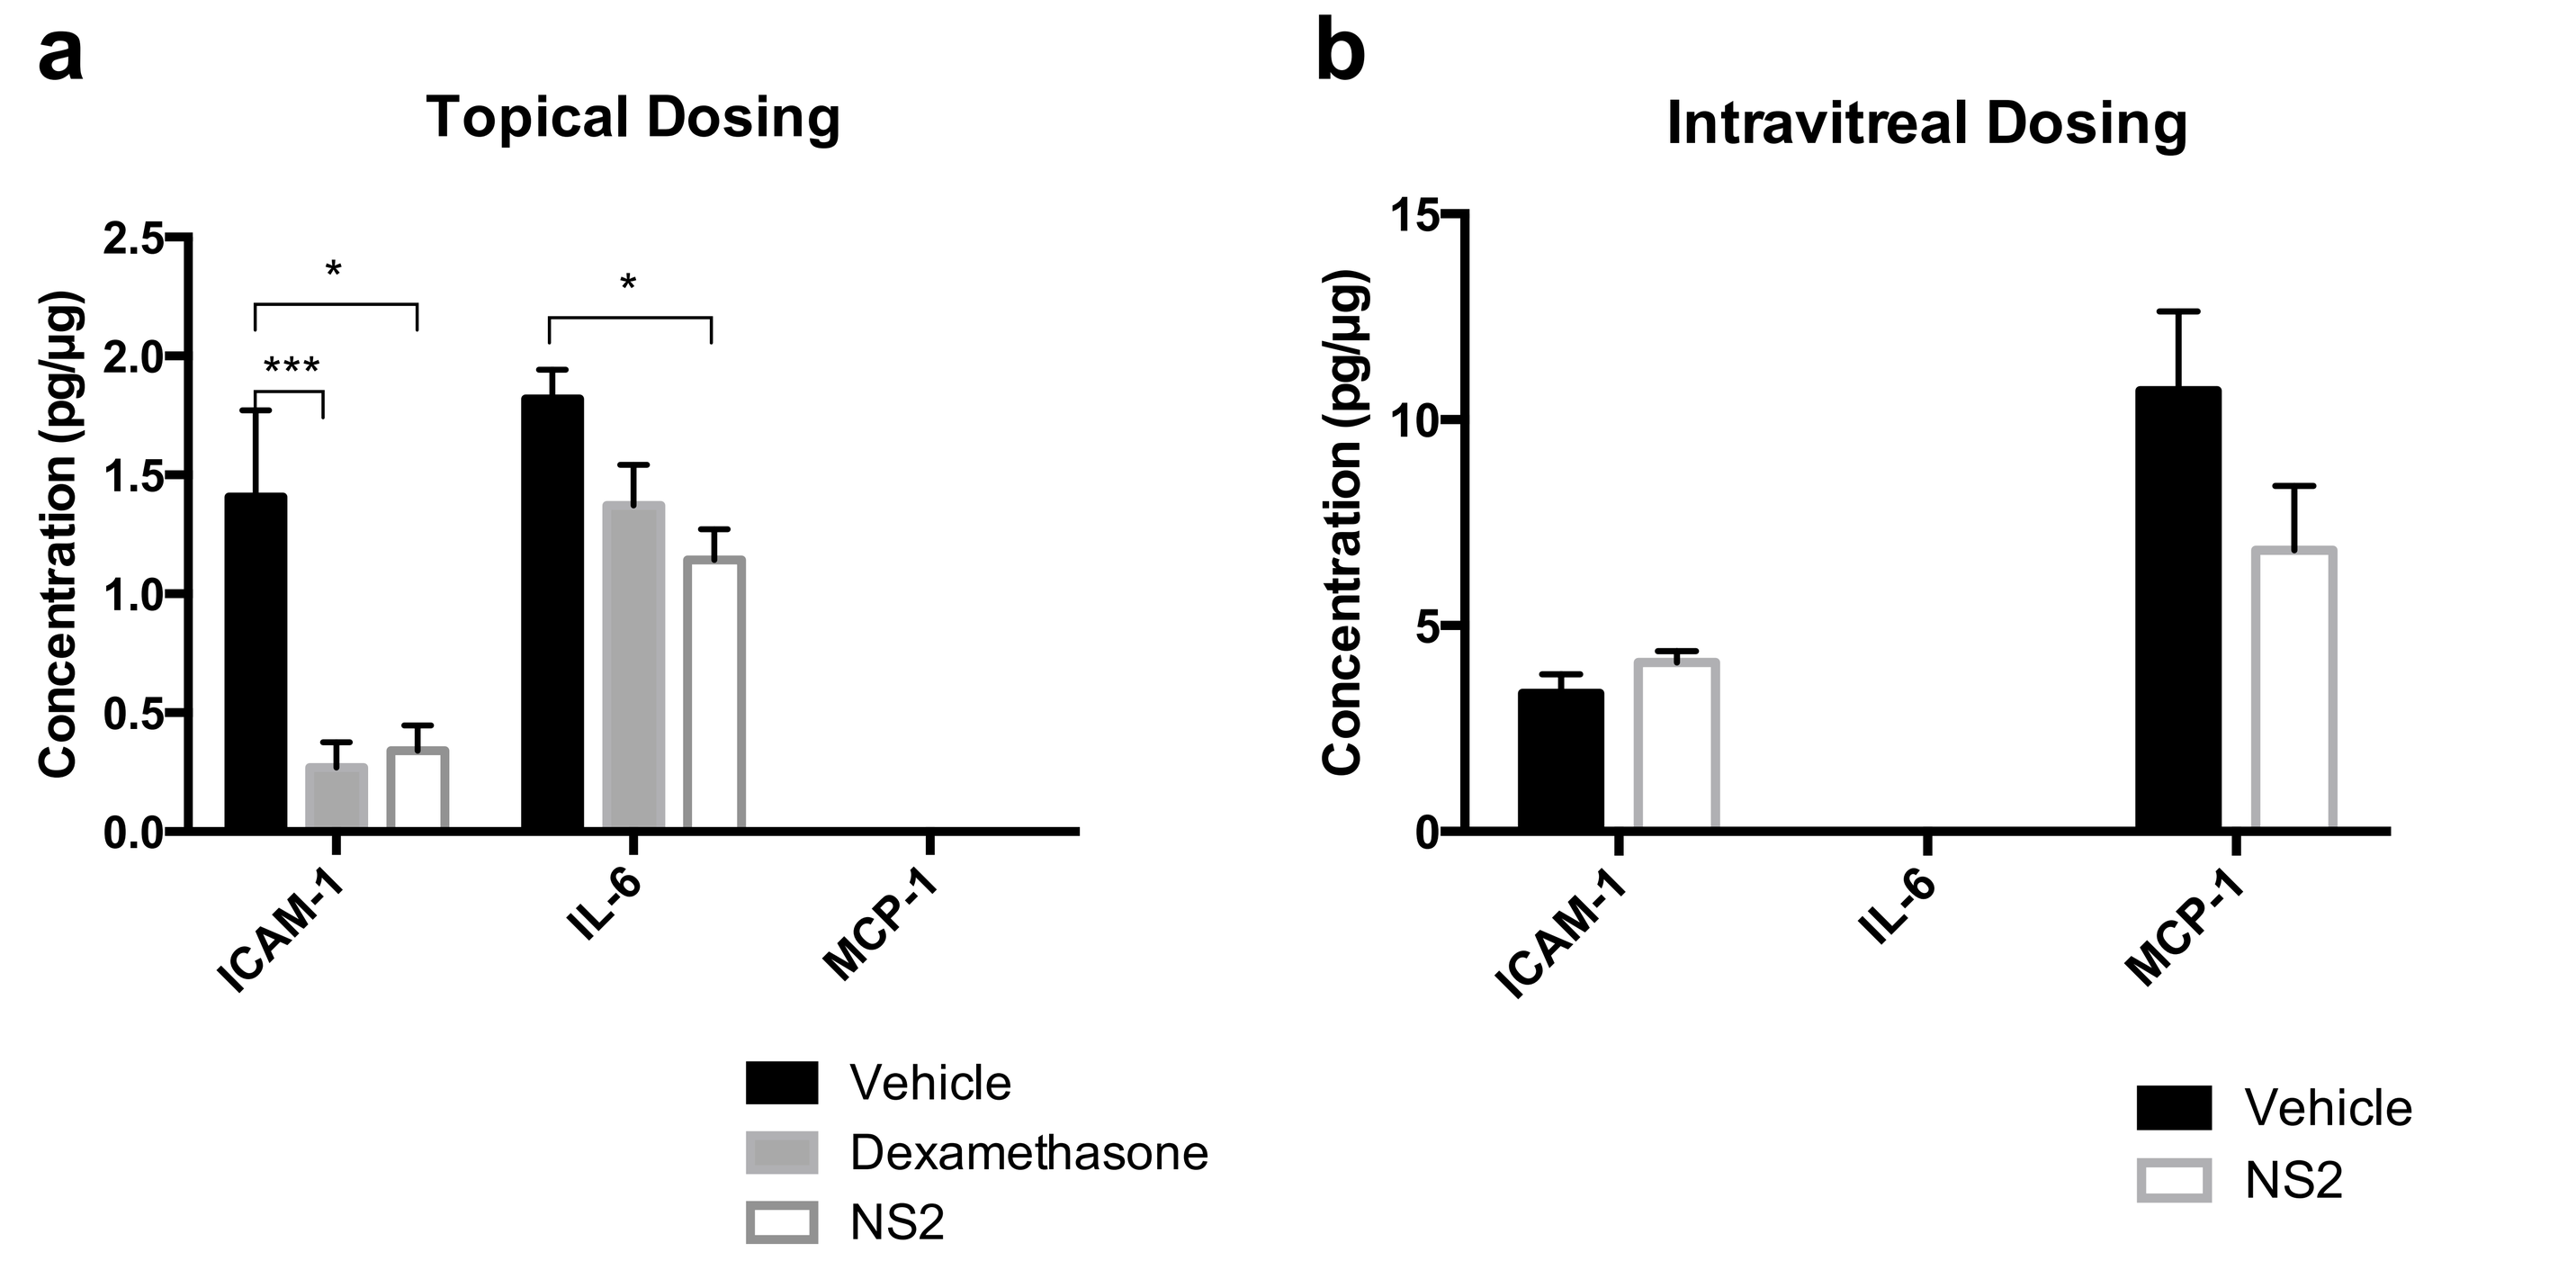
a
Topical Dosing
Concentration (pg/µg)
ICAM-1
IL-6
MCP-1
Vehicle
Dexamethasone
NS2
b
Intravitreal Dosing
Concentration (pg/µg)
ICAM-1
IL-6
MCP-1
Vehicle
NS2

Figure 4. Inflammatory Markers. Levels of inflammatory markers detected in retina-choroid specimens from animals (a) dosed topically with vehicle, dexamethasone or NS2; or (b) dosed intravitreally with vehicle or NS2. * p < 0.05, ** p < 0.01, *** p < 0.001, **** p < 0.0001.